The quick reading method: its efficiency and accuracy in assessing reading performance in the periphery <# 3278 >

THE OHIO STATE UNIVERSITY
COLLEGE OF OPTOMETRY

Laboratory of Vision Enhancement

Shepard, Timothy[1], Hou, Fang[2], Bex, Peter[3], Lesmes, Luis[4], Lu, Zhong-Lin[5], Yu, Deyue[1]

[1] College of Optometry, Ohio State University, Columbus, OH, USA. [2] Wenzhou Medical University, Wenzhou, Zhejiang, CN. [3] Psychology, Northeastern University, Boston, MA, USA. [4] Adaptive Sensory Technology, San Diego, CA, USA. [5] Psychology, Ohio State University, Columbus, OH, USA

Purpose: Patients with central vision loss have to rely on their peripheral vision for reading. Accurate assessment of reading performance can help prescribe suitable adaptive devices to the patients. In this study, we develop an adaptive method, quick reading (qRD), to measure reading speed in the periphery. While the conventional method is adequate, qRD utilizes a Bayesian adaptive framework to select optimal stimuli, thus allowing for an efficient assessment of reading speed in the periphery.

Methods: Seven normally-sighted observers participated. We used a rapid serial visual presentation (RSVP) paradigm where words were serially presented at 10° eccentricity in the lower field. The number of words correctly reported were recorded.

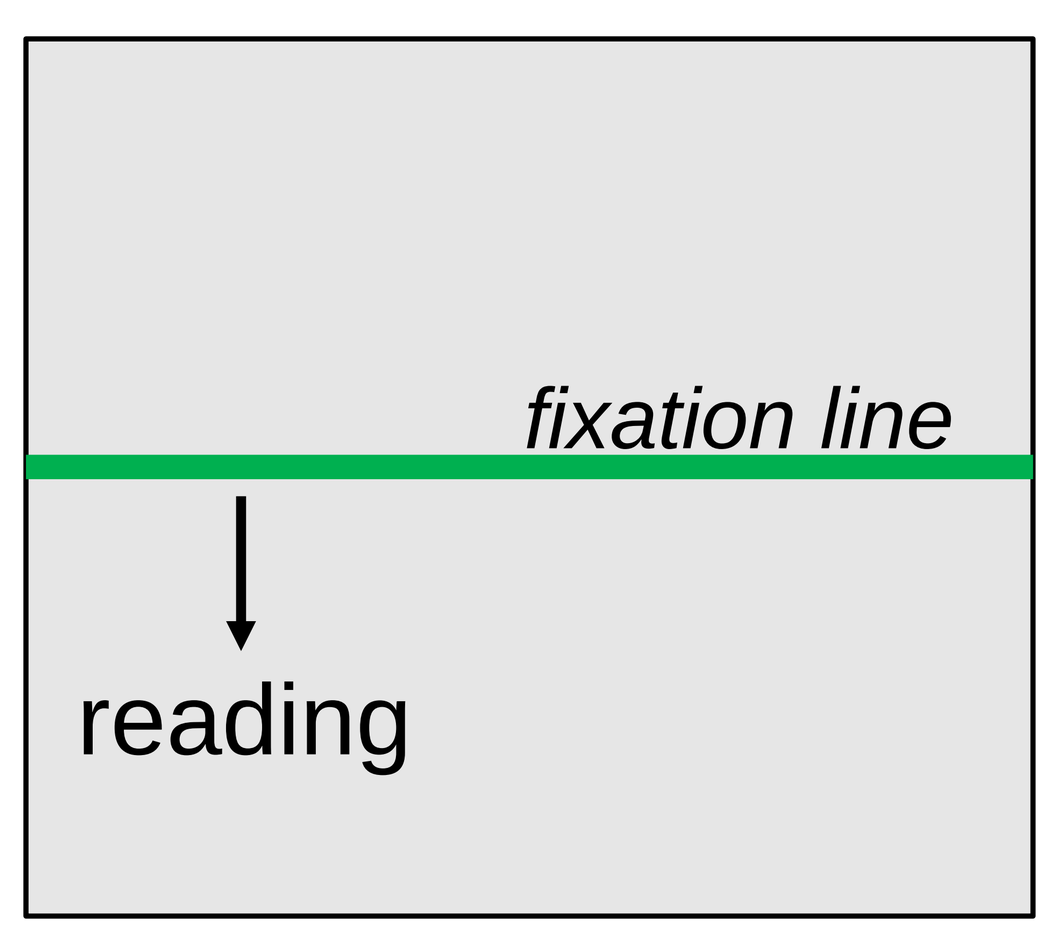
fixation line
reading

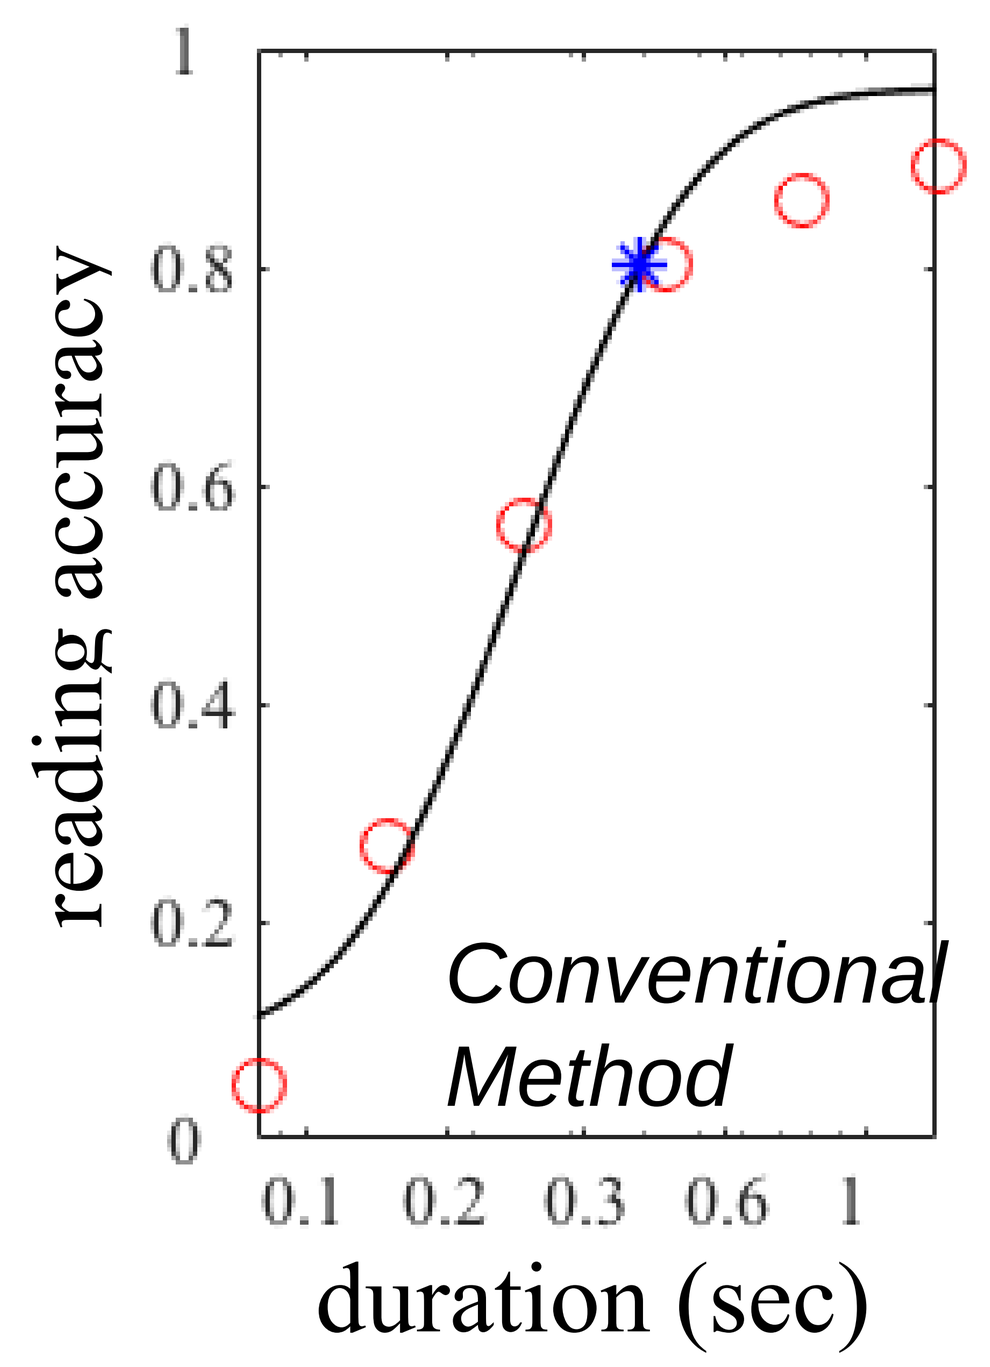
reading accuracy
duration (sec)
Conventional Method
0
0.2
0.4
0.6
0.8
1
0.1
0.2
0.3
0.6
1

RSVP Method

The conventional (method of constant stimuli) procedure involved measuring reading accuracy as a function of exposure duration. Reading speed at a given print size is defined as the duration at which subject's response is 80% correct. The reading speed versus print size function was estimated by measuring reading speed at five print sizes (a total of 180 trials).

Testing with the conventional RSVP method requires ~1 hour of testing for only 5 print sizes.

Quick Reading Method

1) In the qRD procedure, reading speed versus print size was described by an exponential function with three parameters (asymptotic performance level, print size corresponding to a reading speed of 6 wpm, and a decay constant).

2) Following each trial (50 trials total), posterior distributions of the parameters were updated based on subject's response, and stimulus condition (print size and exposure duration) was selected to provide the maximal expected information gain for the upcoming trial.

The Quick Reading method requires ~12 mins of testing and also allows for a large array of possible test stimuli (e.g. 25 print sizes x 50 exposure durations).

Results: Reading curves (reading speed vs. print size) estimated using the two methods were comparable across observers (area under curve: $t(6)=2.44$, $p=0.72$). The conventional data was analyzed using the Bayesian fitting component of qRD. A paired-samples t-test was conducted to compare 68.2% credible intervals between the qRD and conventional methods. The qRD method was more precise (i.e. smaller credible intervals) than the conventional method when considering only 50 conventional trials ($p=0.01$) and similar when 180 conventional trials were included ($p=0.50$).

Conclusions: The current investigation demonstrates that the qRD method can adequately measure reading function in the periphery but with higher precision than the conventional method when only 50 conventional trials are considered.

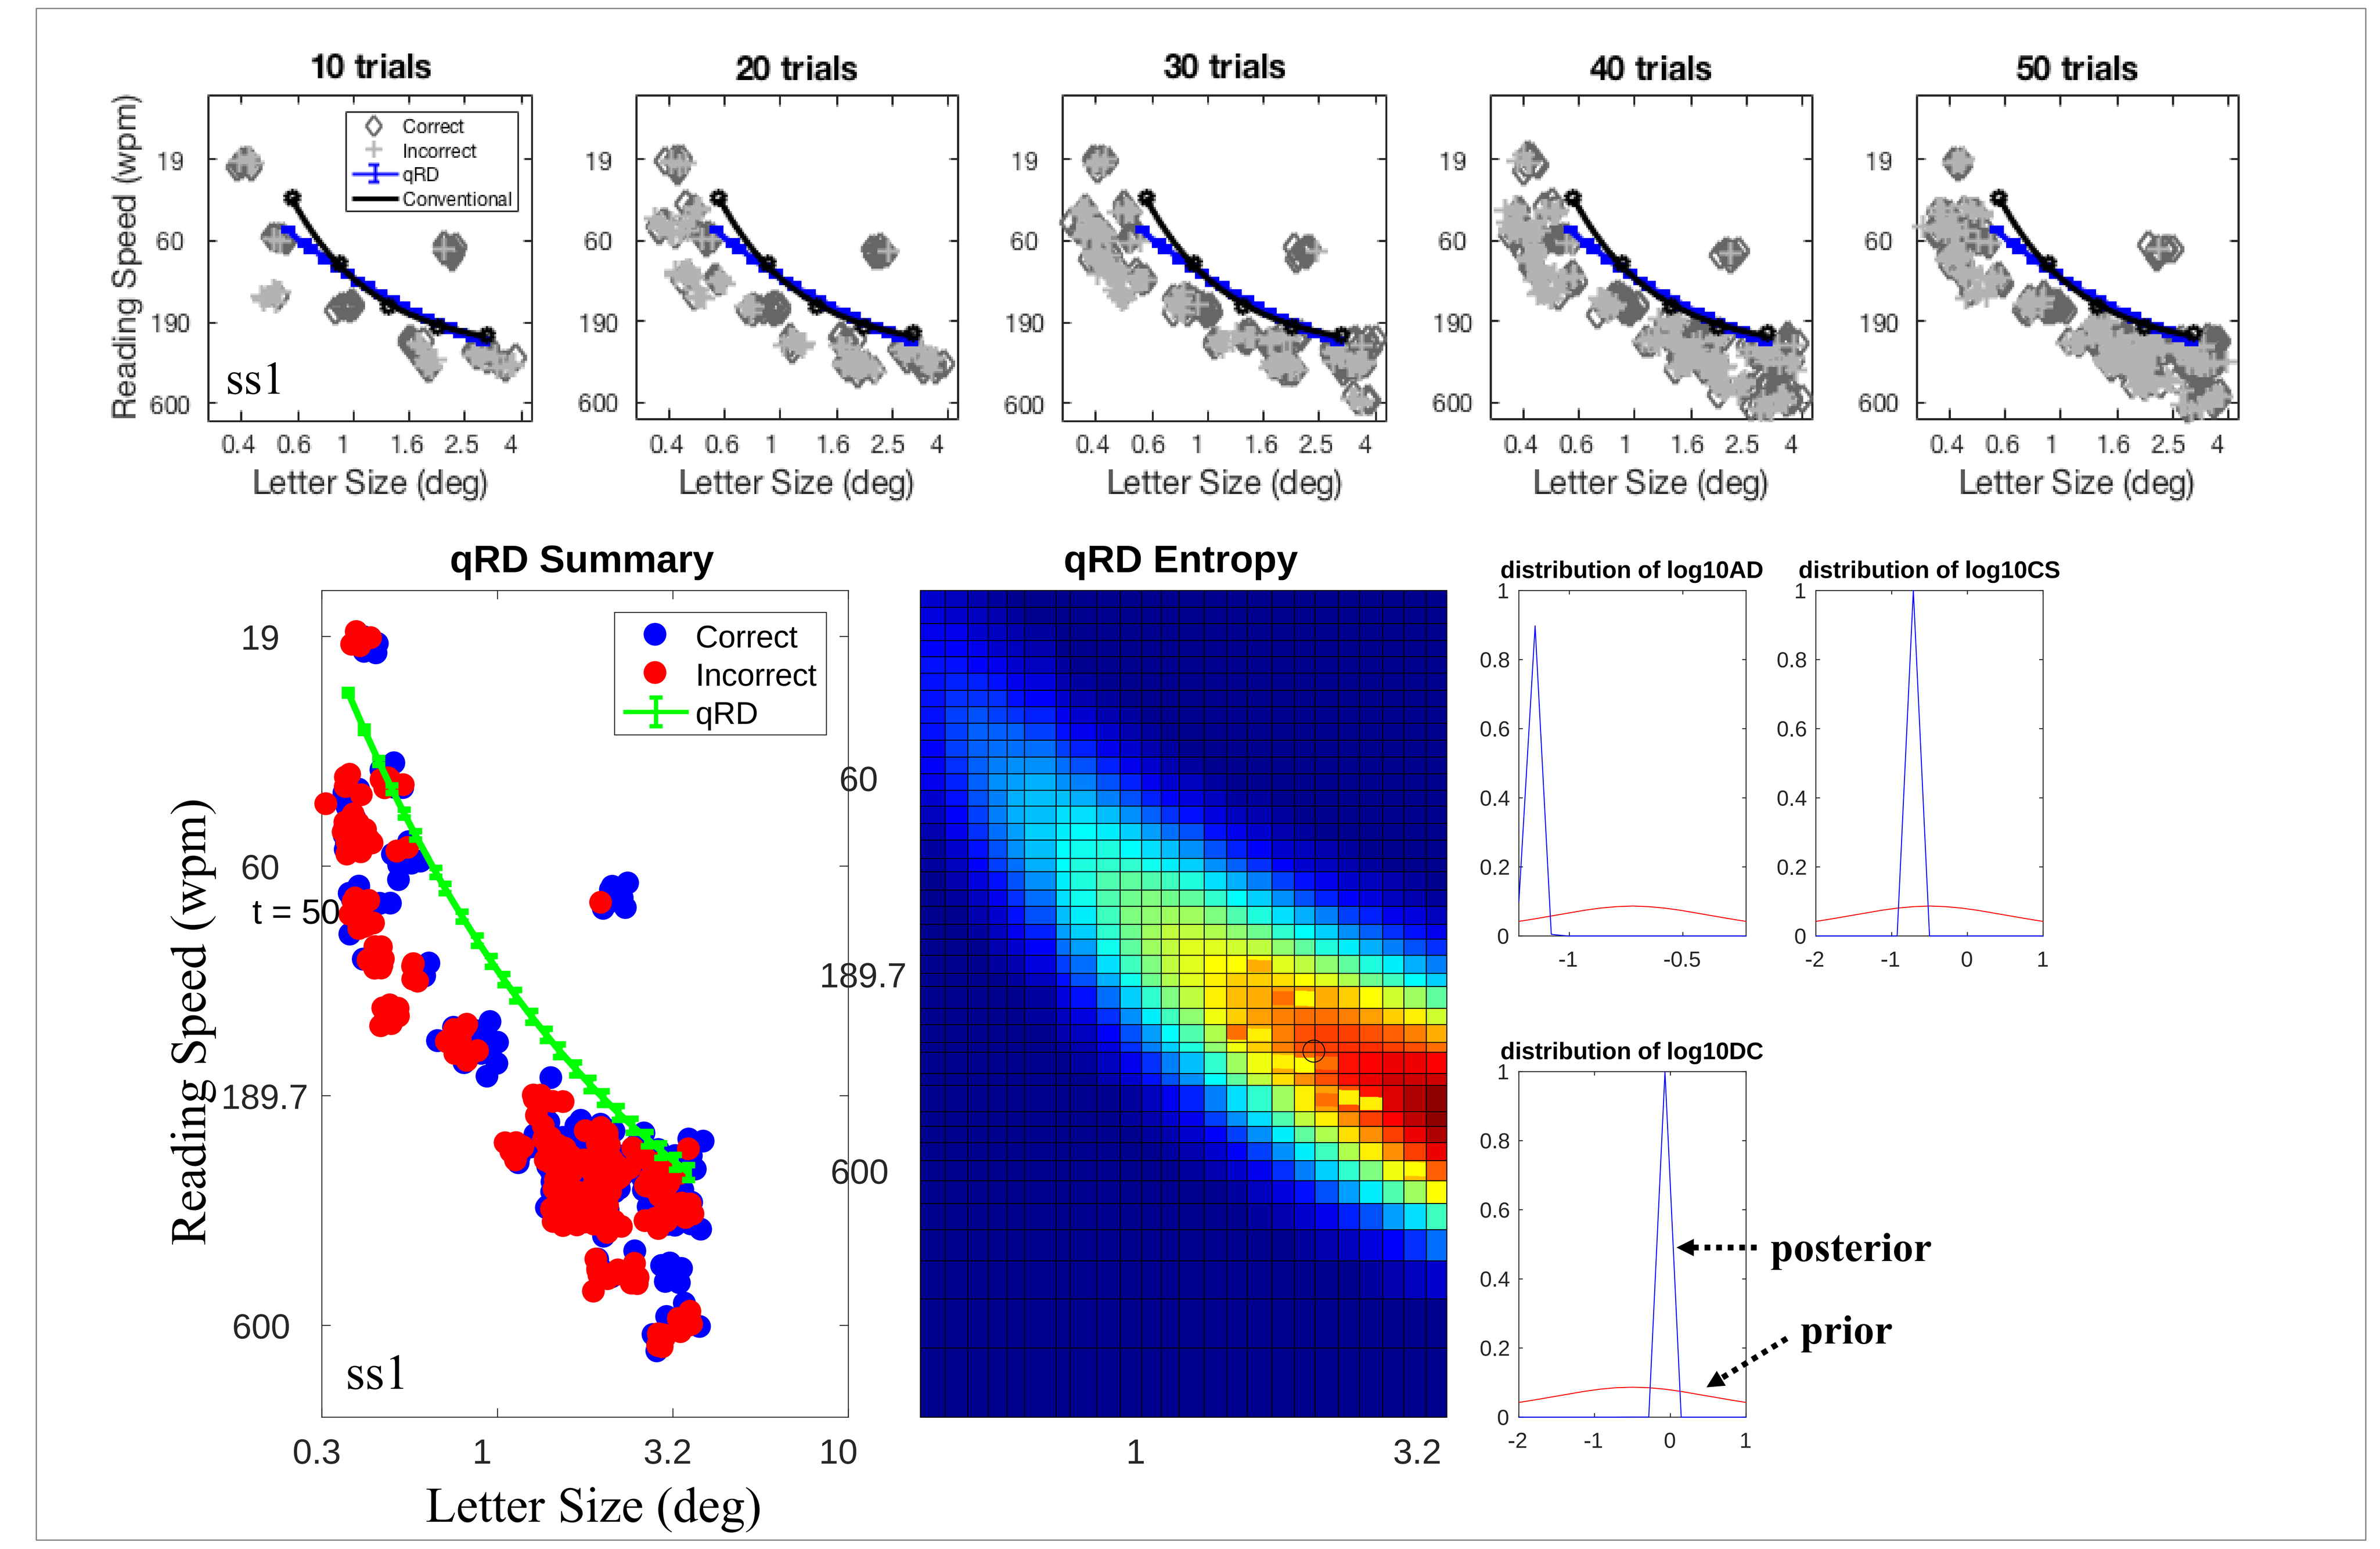
10 trials
20 trials
30 trials
40 trials
50 trials
Reading Speed (wpm)
Letter Size (deg)
Correct
Incorrect
qRD
Conventional
ss1
qRD Summary
qRD Entropy
t = 50
distribution of log10AD
distribution of log10CS
distribution of log10DC
posterior
prior

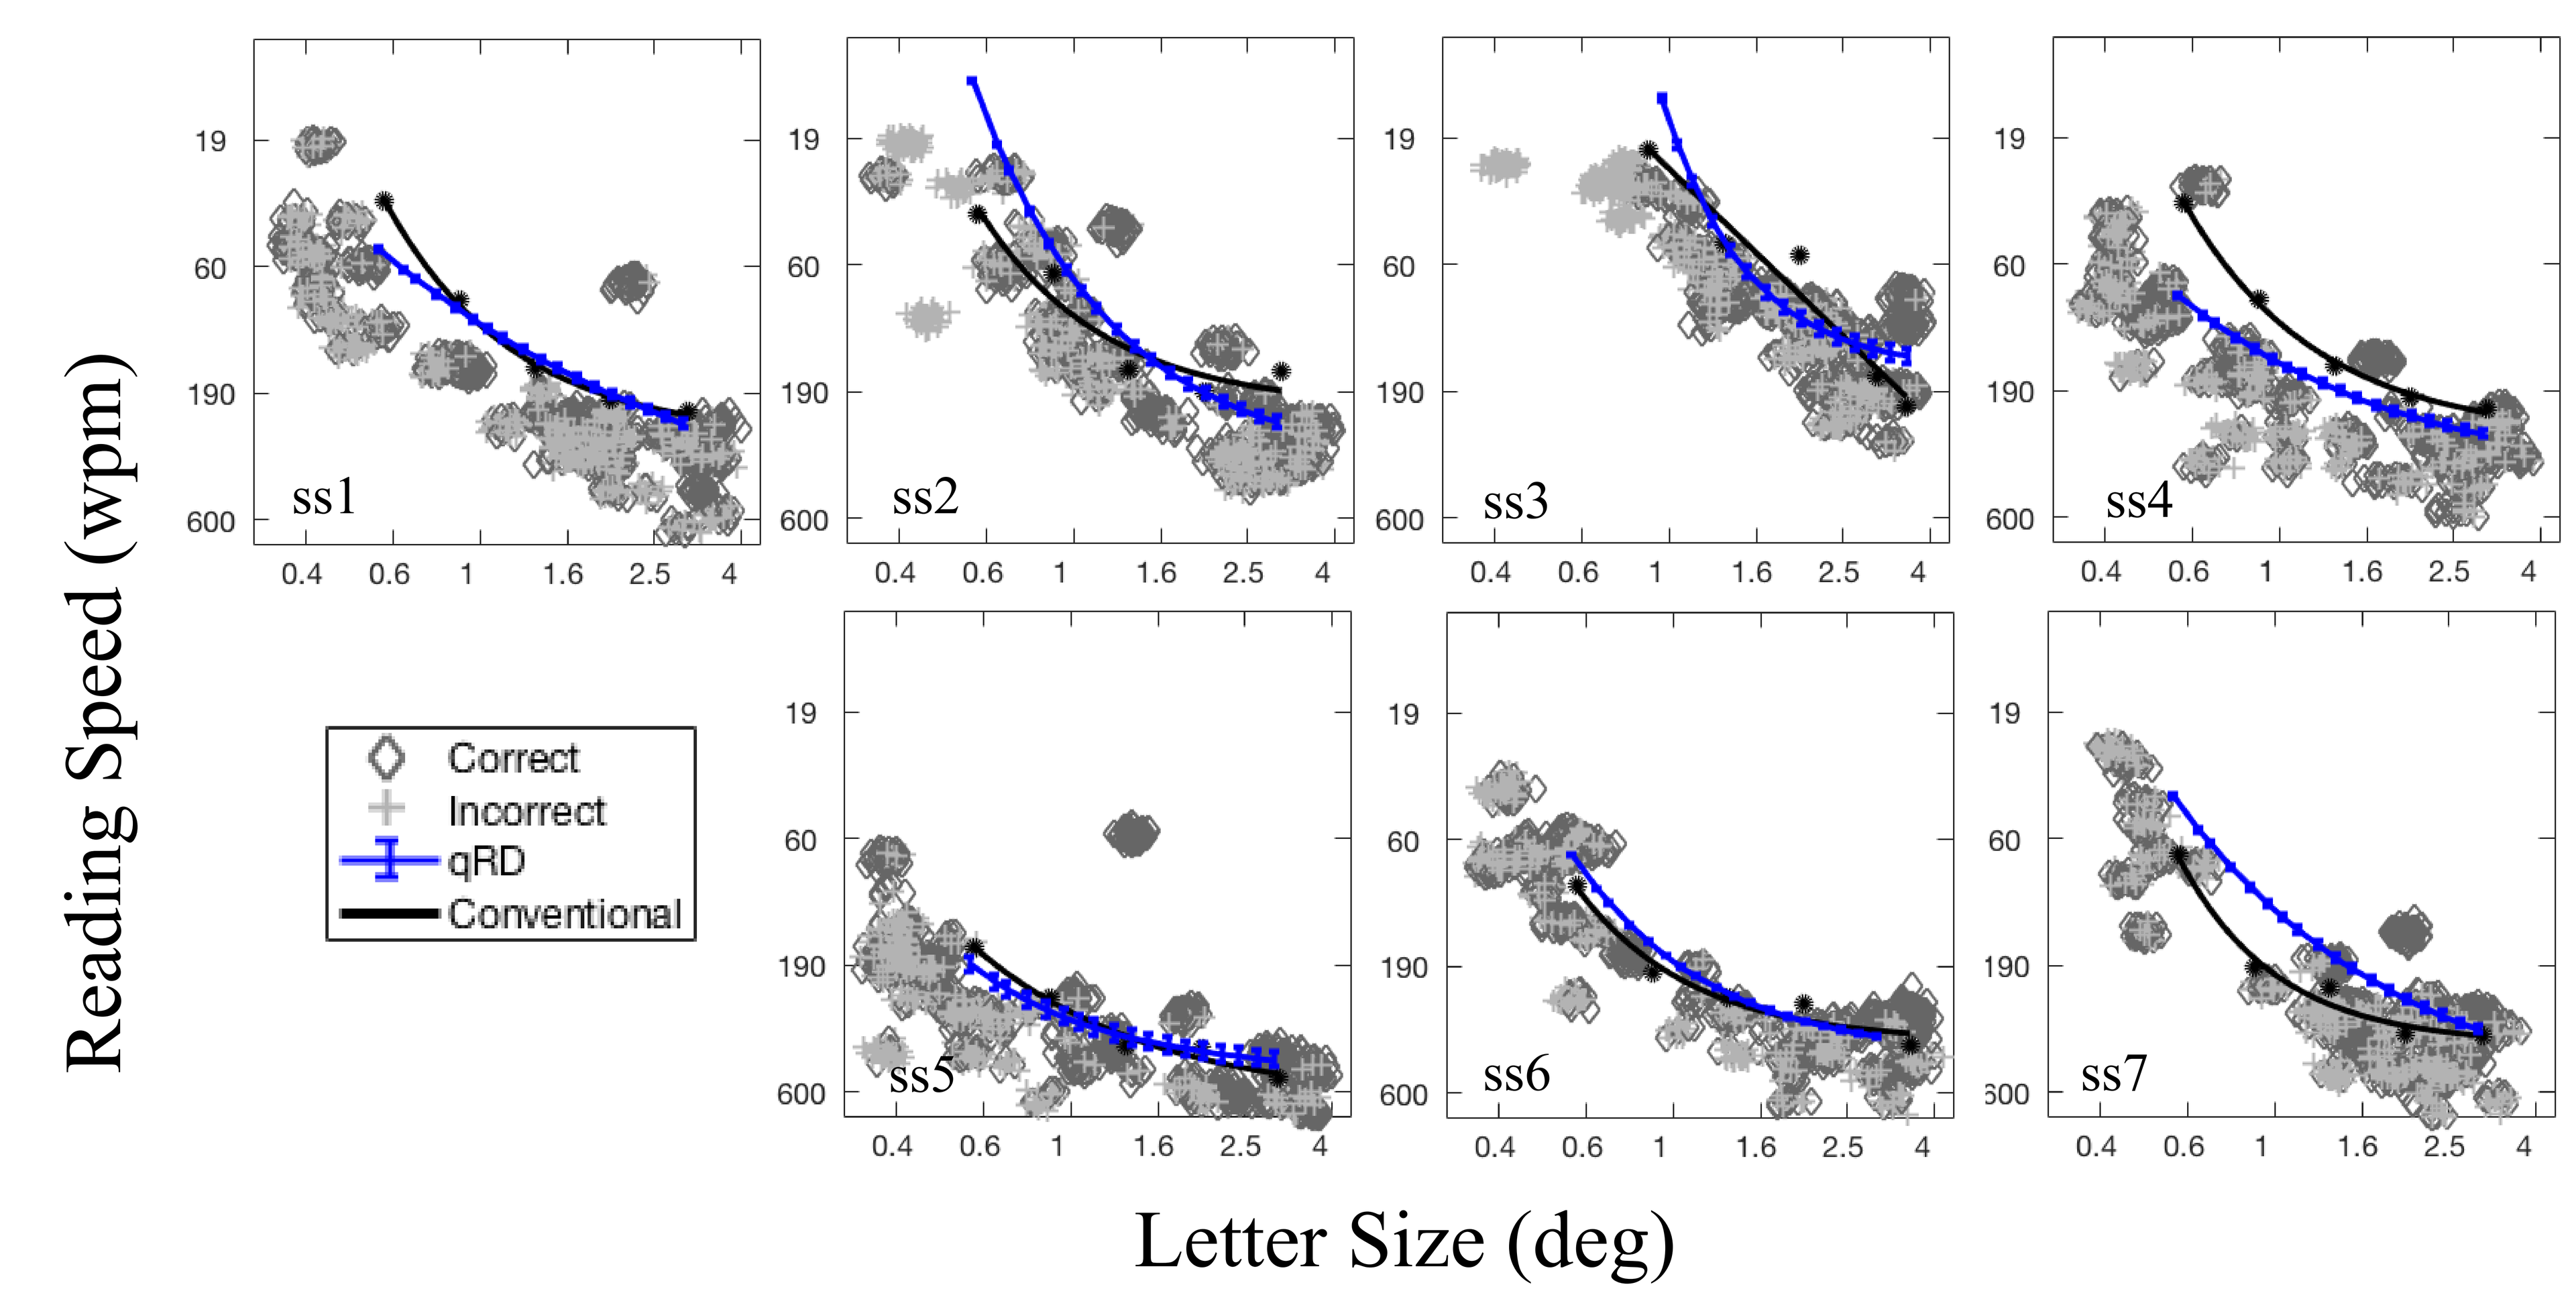
Reading Speed (wpm)
Letter Size (deg)
ss1
ss2
ss3
ss4
ss5
ss6
ss7
Correct
Incorrect
qRD
Conventional

Acknowledgements: NIH grants EY025658(DY) and EY021553 (ZLL) Timothy Shepard: No Commercial Relationship (CR): Code N | Fang Hou: CR;US Provisional Patent 62/378,334: Code P | Peter Bex: CR; Adaptive Sensory Technology (AST): Code I; ASTP | Luis Lesmes: CR;AST; AST: Code P; AST: Code E | Zhong-Lin Lu: CR; AST: Code P; AST: Code I | Deyue Yu: CR;US Provisional Patent 62/378,334: Code P