2904 - D0003

Compensatory strategies for independent binocular scotomas in simulated CFL

Concetta F. Alberti, Peter J. Bex

Introduction

Binocular Central Field Loss (bCFL) is the loss of central vision in both eyes.
Without a binocular target the eyes tend to lose fusion.

Vergence anormalities caused by the binocular scotomas are used 'strategically' without specific instructions.

Methods

bCFL simulation: 144 Hz shutter glasses presenting a Gaze-Contingent (GC) Blob in each eye.
GC Blob: Gaussian pink noise with 1.12 – 2.24 – 4.48 – 8.96 degrees std. Blob size varied between-blocks.
Task: 50 trials Single-Letter Acuity task with letter size controlled through a staircase method.
Participants: 11 NV -two experts, 9 naïve.

Hypothesis

- The larger the scotoma size the greater the vergence anormalities.

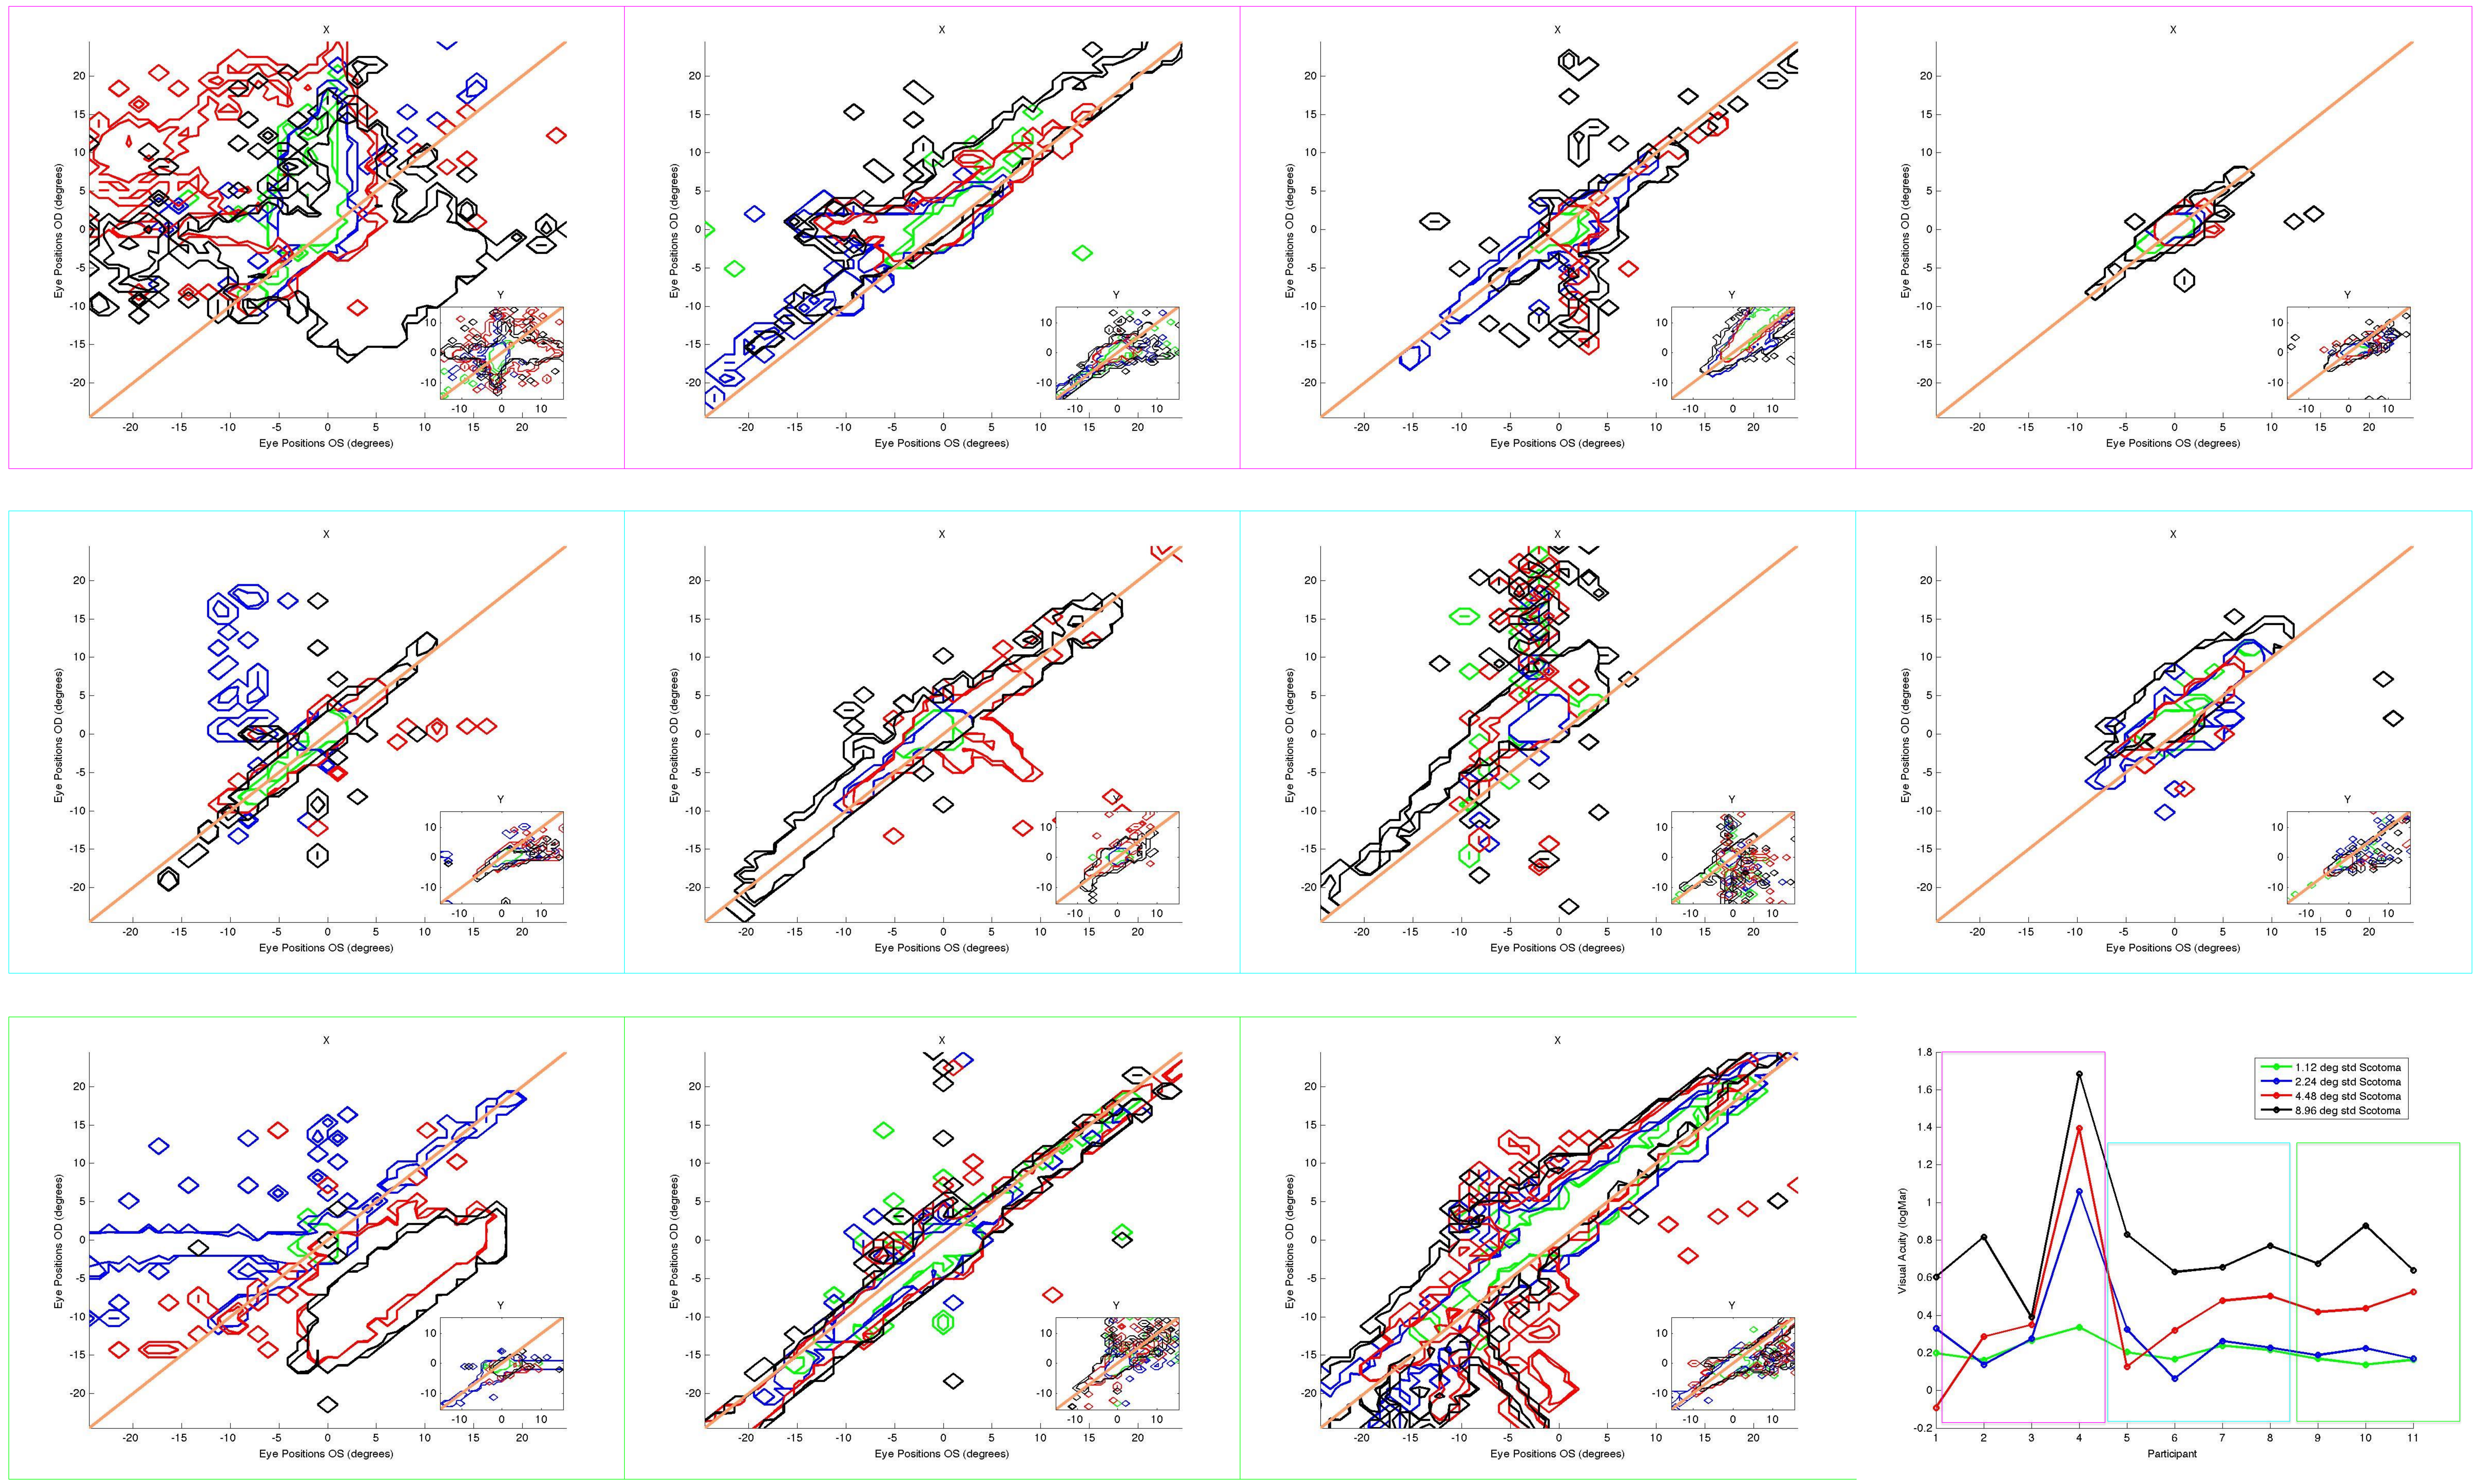
X
Y
Eye Positions OD (degrees)
Eye Positions OS (degrees)
1.12 deg std Scotoma
2.24 deg std Scotoma
4.48 deg std Scotoma
8.96 deg std Scotoma
Visual Acuity (logMar)
Participant